COMPARISON OF SMALL VIRUS CLEARANCE AND FLOW RATES BETWEEN MEISSNER'S SEPRAPOR® HOLLOW FIBER ULTRAFILTRATION 50 KDA AND 500 KDA FILTER MEMBRANES USING ΦX174 BACTERIOPHAGE

Zachary Bendiks, Ph.D., Mao Kohara, Leesa McBurnie

Abstract

Viral clearance by virus-retentive filters is a crucial step in many biomanufacturing process streams, including production of monoclonal antibodies and cell culture media. Studies with model bacteriophages or viruses are needed to determine whether a specific filter should be categorized as a small or large virus-retentive filter. In this study, viral retention and flow rates were compared between Meissner's 50 kDa and 500 kDa SepraPor® Hollow Fiber (HF) Ultrafiltration (UF) membranes used for Tangential Flow Filtration (TFF). These filters were challenged with ~10 million plaque forming units (PFU) of the small bacteriophage ΦX174 suspended in deionized (DI) water. It was determined that the 50 kDa SepraPor® HF UF membranes gave log-reduction values (LRVs) ≥ 5 with flow rates ranging from 15.9 to 46.7 mL/min at 15 psi constant pressure. The 500 kDa SepraPor® HF UF membranes gave LRVs ≤ 0.74 with flow rates ranging from 8.8 to 286 mL/min. These results show that Meissner's 500 kDa SepraPor® filter membrane does not retain ΦX174 in water and does not function as a small virus retentive filter, while the 50 kDa SepraPor® filter membrane gives robust clearance of ΦX174 and can be categorized as a small virus retentive filter.

Material & Methods

Meissner SepraPor® UF HF 50 and 500 kDa TFF filters, tubing, gaskets, clamps, valves, reservoir, DI water, and collection bottles were autoclaved at 121°C for 60 minutes then placed in a BSC. The challenge solution was prepared by diluting a stock aliquot of the *E. coli* bacteriophage ΦX174 (ATCC 13706-B1™) in 1 L of sterile DI water. A pressure source was attached to the upstream side of the reservoir and the test filter was connected to the outlet. For each filter, approximately 200 mL challenge solution was added to the reservoir. The filter was vented at < 5 psi until challenge solution escaped through the retentate valve, then the valve was closed and the permeate tubing clamp opened. Pressure was increased to 15 psi and the permeate collected in a glass bottle. A timer was started once the first drop of challenge solution was collected from the permeate port. Pressure was turned off and the timer stopped once the flow through the permeate slowed. Challenges were performed on triplicate capsules for each pore size. As a control, 0.2 µm disc filters were tested in parallel with the HF UF filters. All challenges were performed in a BSC at room temperature. Each test filter was integrity tested before and after autoclaving and after the challenge using the diffusive flow test in water with air at 30 psi, while the control discs were integrity tested with the bubble point test in water.

Viral titers in each filter permeate were quantified using the plaque assay. Ten-fold serial dilutions were performed for each permeate sample. For each dilution, 1 mL was added to a Nutrient Agar (NA) plate. 100 µL of log-phase *E. coli* (ATCC 13706™) grown in Nutrient Broth (NB) was mixed with 2.5 mL of Nutrient Soft Agar, pre-warmed to 50°C, and this mixture was poured onto the NA plate and lightly rotated to cover the plate surface. Plates were incubated overnight at 37°C and counted the following day. Log-reduction values (LRV) were determined by dividing the number of PFU each filter encountered by the number of PFU in the permeate, then calculating the Log base 10 of this ratio.

Pressure source at 15 psi

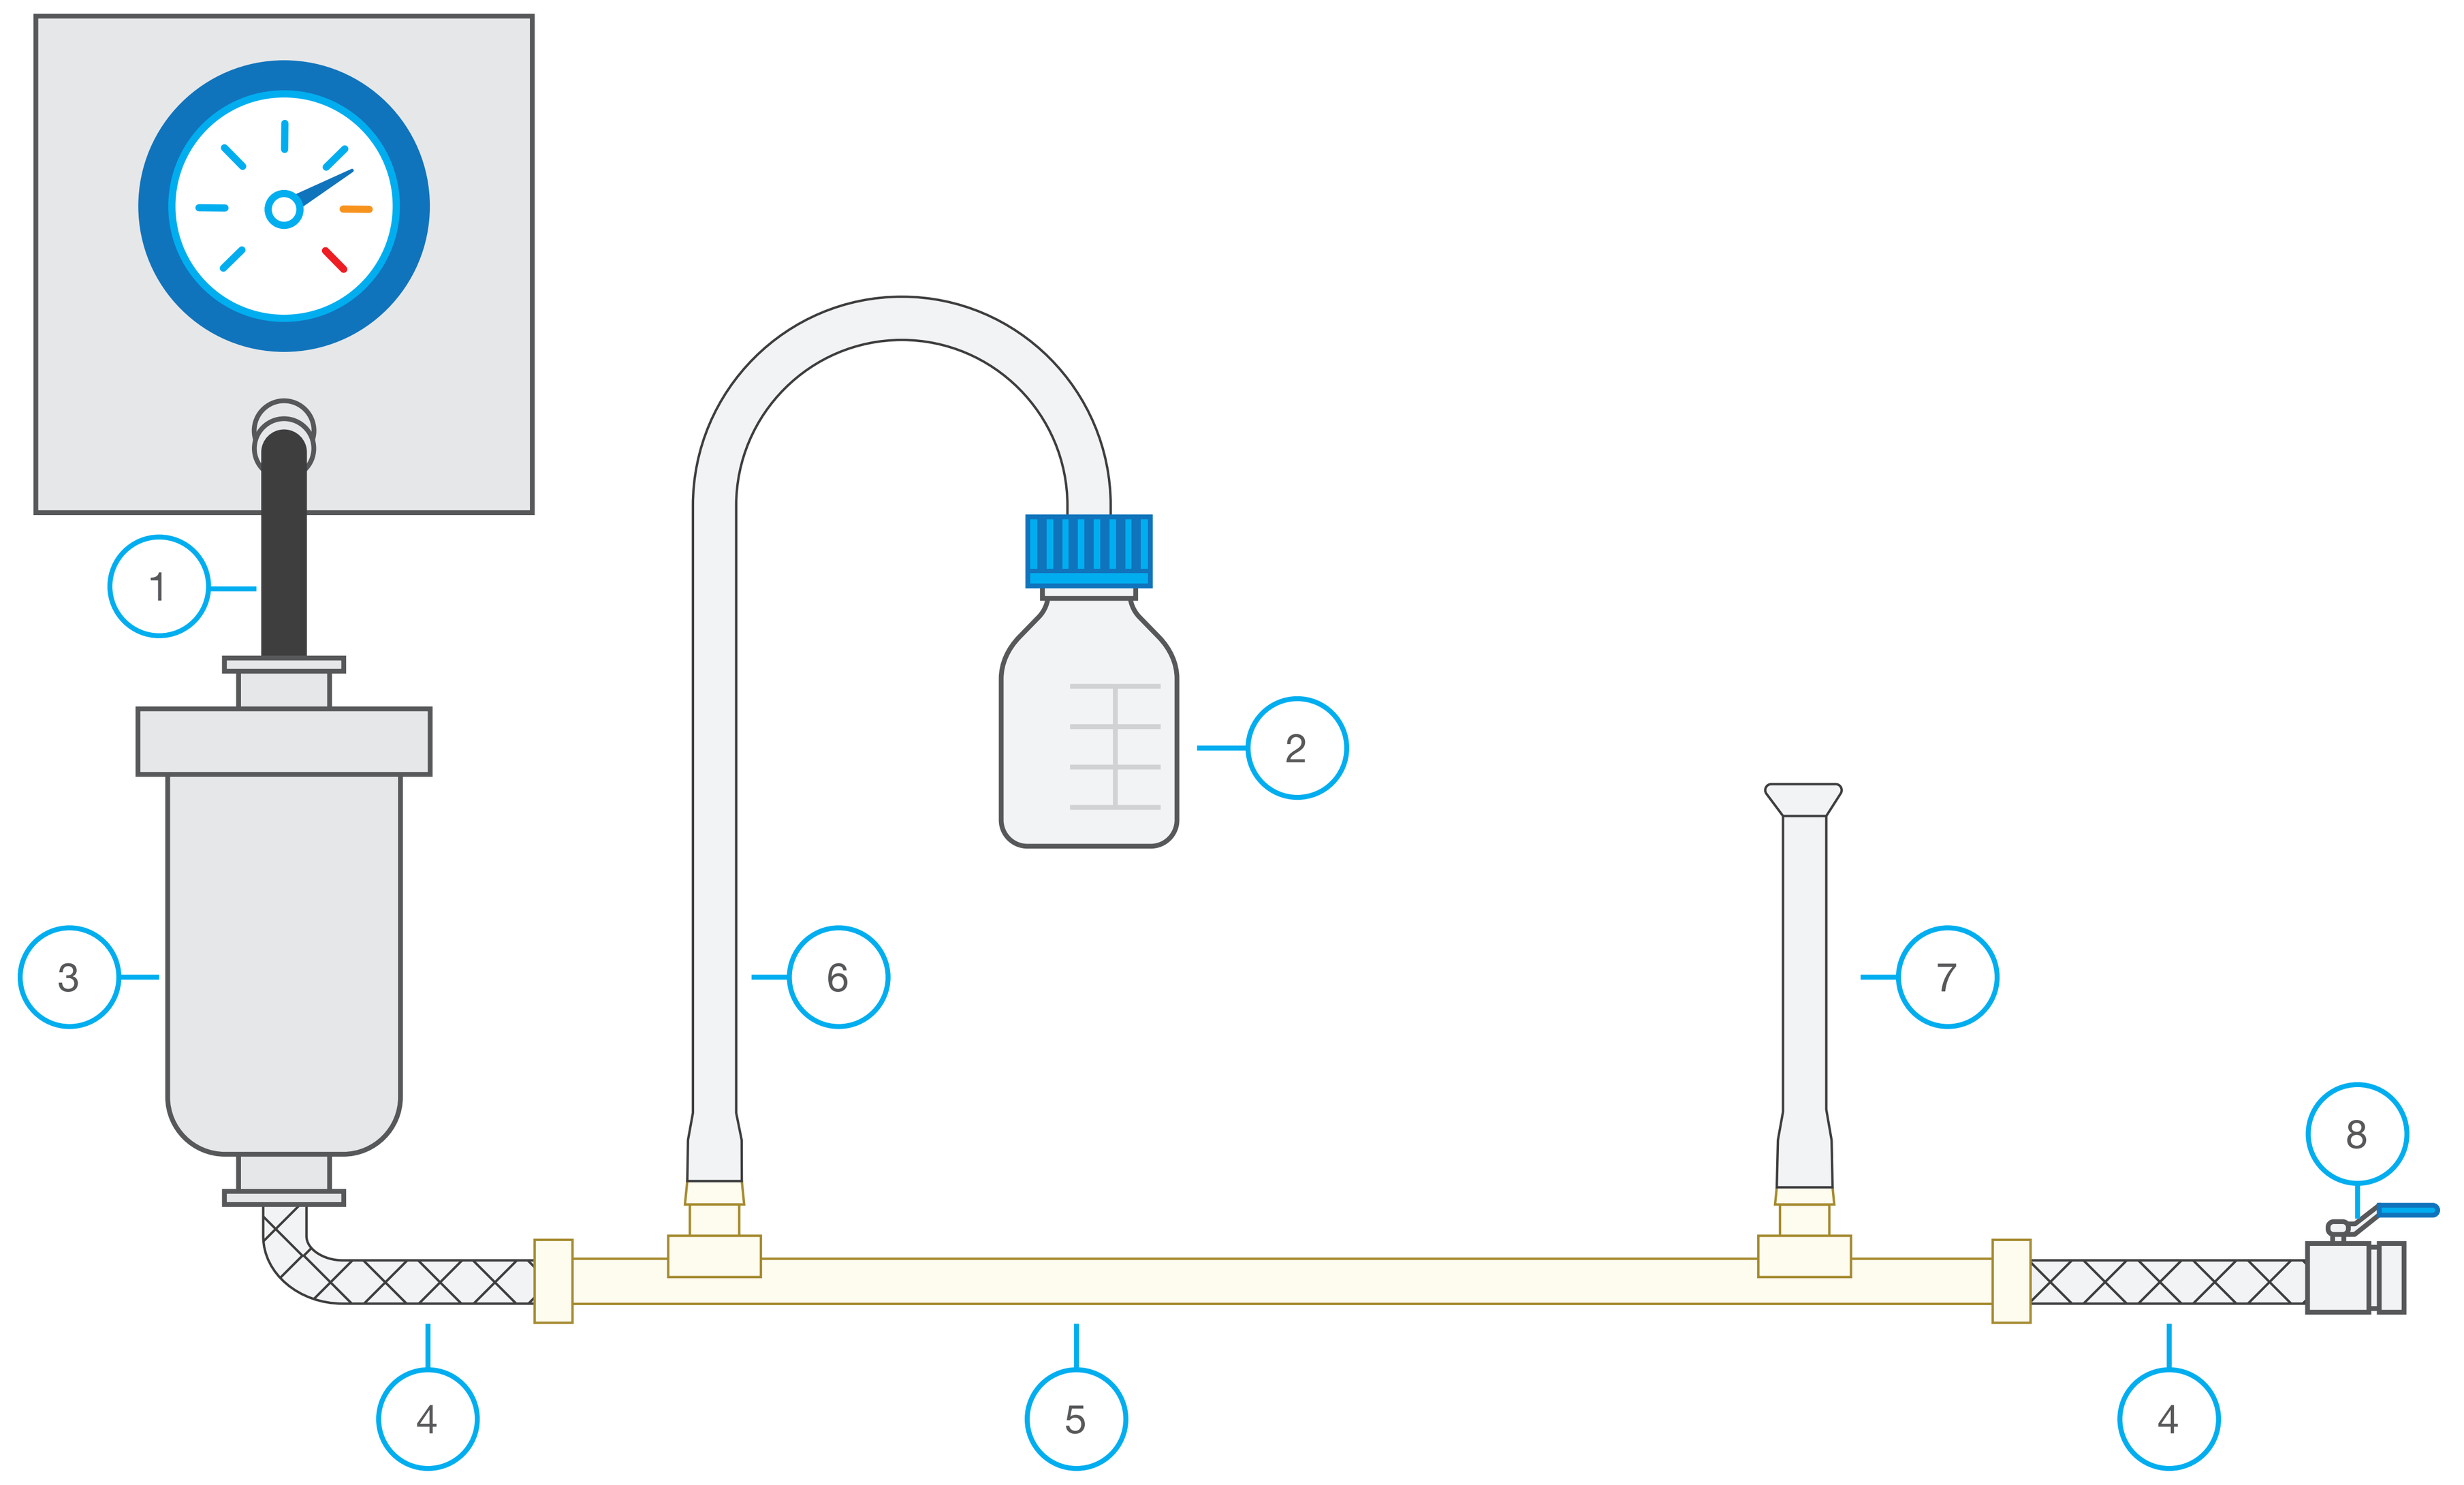
1
2
3
4
5
6
7
8

1. Polyethylene tubing
2. Permeate collection bottle
3. Sanitary Inline Filter Housing Reservoir
4. M-Sil™ Braid-reinforced silicone tubing
5. SepraPor® TFF Filter 50 or 500kDa
6. Silicone tubing
7. Silicone tubing, clamped
8. Retentate Valve

Results

Filter number	PFU Challenged	PFU in Permeate
SepraPor® 50 kDa UF HF Membrane		
1	1.39×10^7	140
2	1.73×10^7	0
3	1.29×10^7	0
SepraPor® 500 kDa UF HF Membrane		
1	2.60×10^7	6.30×10^6
2	5.20×10^7	9.40×10^6
3	9.10×10^6	2.14×10^6
0.2 µm PVDF Disc Filter (Control)		
1	2.28×10^7	2.37×10^7
2	4.68×10^7	4.50×10^7

ΦX174 bacteriophage challenge levels and the number of PFU remaining in the permeate for SepraPor® 50 and 500 kDa UF HF membrane filters used for TFF and for the 0.2 µm PVDF control disc filters.

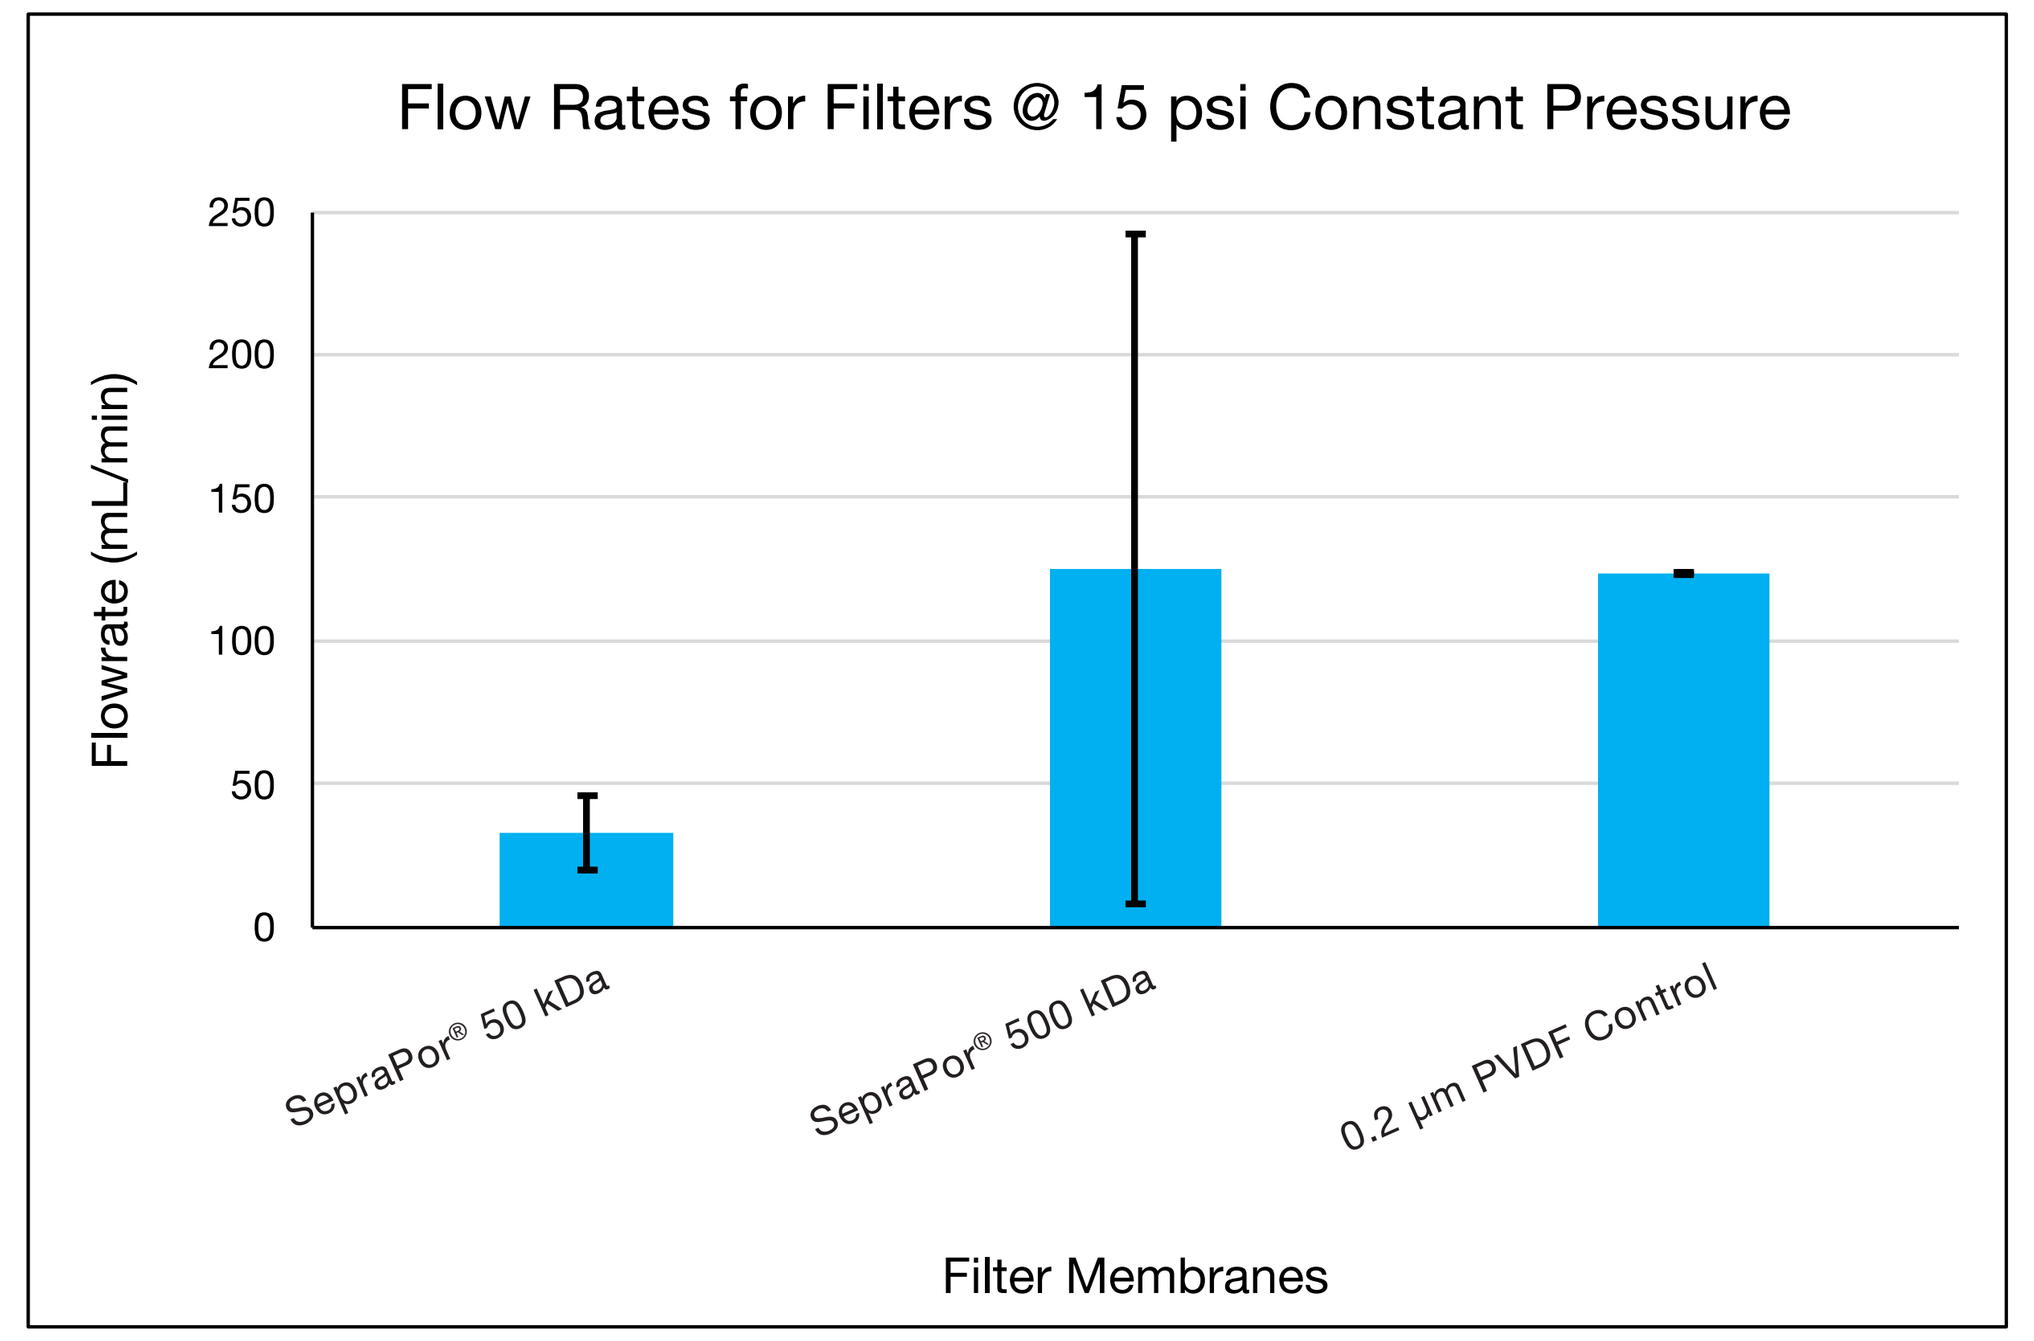
Flow Rates for Filters @ 15 psi Constant Pressure
Flowrate (mL/min)
250
200
150
100
50
0
SepraPor® 50 kDa
SepraPor® 500 kDa
0.2 µm PVDF Control
Filter Membranes

Measured flow rates of 200 mL DI water containing ~10^5 PFU/mL ΦX174 bacteriophage through the permeate ports of SepraPor® 50 and 500 kDa UF HF membrane filters or through the 0.2 µm PVDF control disc filters.

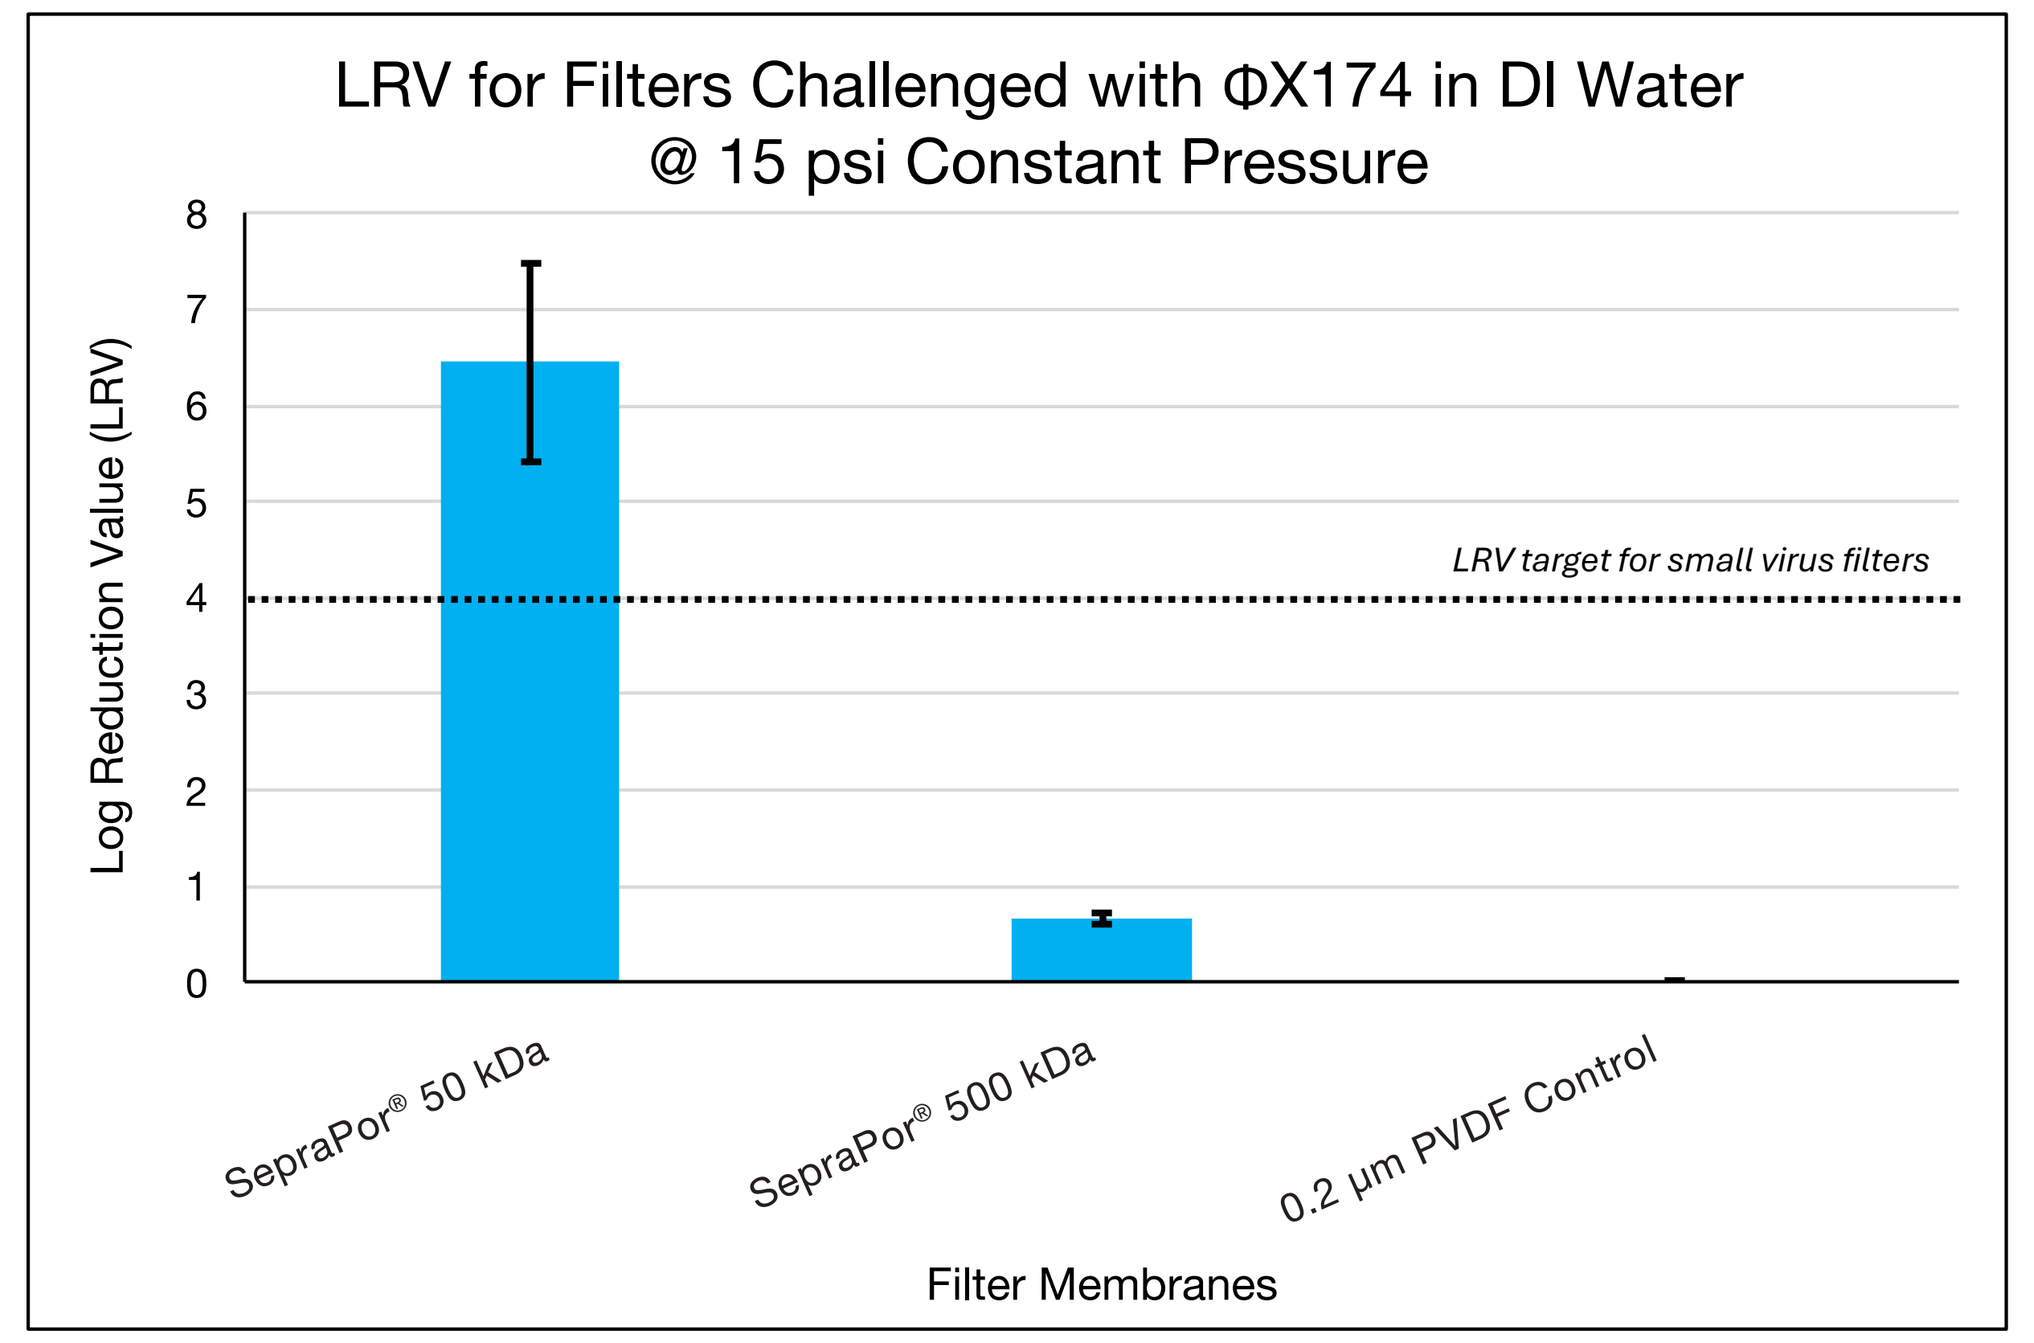
LRV for Filters Challenged with ΦX174 in DI Water
@ 15 psi Constant Pressure
Log Reduction Value (LRV)
8
7
6
5
4
3
2
1
0
LRV target for small virus filters
SepraPor® 50 kDa
SepraPor® 500 kDa
0.2 µm PVDF Control
Filter Membranes

LRV's of SepraPor® 50 and 500 kDa UF HF membrane filters and the 0.2 µm PVDF control disc filters when challenged with 200 mL DI water containing ~10^5 PFU/mL ΦX174 bacteriophage.

Conclusion

In this study, flow rates and viral retention were characterized for Meissner's SepraPor® 50 and 500 kDa UF HF membranes used for TFF when challenged with the small *E. coli* bacteriophage ΦX174 in DI water at 15 psi constant pressure. The SepraPor® 50 kDa membrane produced LRV's ranging from 5.00 to > 7.24, with two filters producing sterile filtrates, and measured flow rates of 16 to 47 mL/min. The SepraPor® 500 kDa membrane produced LRV's ranging from 0.62 to 0.74 and measured flow rates of 8.8 to 286 mL/min. The SepraPor® 50 kDa membrane gave robust viral clearance of ΦX174 and can be used as a small virus filter while the 500 kDa cannot.

www.meissner.com
For additional information, please contact: info@meissner.com

MEISSNER