KEY FACTORS IMPACTING VIRAL RETENTION, PROTEIN PASSAGE & FLOW RATES IN ULTRAFILTRATION

Zachary Bendiks, Ph.D.[1] • Vanessa Santos[1] • Leesa McBurnie[1]

[1]Meisser Corporation, Camarillo, CA

Abstract

Viral filtration by ultrafiltration membranes is a critical step to ensure viral removal in many biomanufacturing process streams. However, the nature of the feed solution and the filtration parameters used can significantly affect filter performance, product quality, and safety. In this work, the impact of three different parameters on ultrafiltration performance was investigated: 1) feed concentration, 2) protein size, and 3) inlet pressure. Findings indicate that the protein concentration of the feed solution affects flow rates in a dose-dependent manner but did not significantly alter retention of ΦX174 bacteriophage particles at the concentrations tested. Results further demonstrated that flow rates and downstream recovery of low-MW BSA were significantly higher than high-MW HgG at the same feed concentration, indicating that the molecular weight of the protein solution significantly alters ultrafiltration performance and downstream product recovery. Finally, testing showed that increasing the inlet pressure increased flow rates and throughput but decreased downstream protein recovery. Therefore, increased pressure has benefits and drawbacks that must be carefully evaluated when developing ultrafiltration process parameters.

Material & Methods

A commercially available ultrafiltration (UF) filter was used for all experiments. The UF filter, tubing, gaskets, clamps, valves, reservoir, and collection bottles were autoclaved at 121°C for 60 minutes prior to the start of each experiment. Protein solutions were prepared by dissolving human gamma globulin (HgG) or bovine serum albumin (BSA) in 1x PBS at the concentrations indicated then sterile-filtering each solution through a 0.2 µm disc filter. To perform the filtration experiments, a pressure source was attached to the upstream side of the reservoir, and the test filter was connected to the outlet. Feed solution was added to the reservoir, the filter was vented at < 5 psi until feed solution escaped through the retentate valve, then the valve was closed and the permeate tubing clamp opened. Pressure was increased to the indicated pressure, and the permeate collected in a glass bottle on a scale to measure throughput over time. Each test filter was integrity tested before and after autoclaving, and after the challenge to ensure they remained integral.

For viral retention testing, the challenge solution was prepared by diluting a stock aliquot of the *E. coli* bacteriophage ΦX174 (ATCC 13706-B1™) in the sterile-filtered protein solution. Following ultrafiltration, viral titers in each filter permeate were quantified using the plaque assay. Ten-fold serial dilutions were performed for each permeate sample. Each dilution was mixed with log-phase *E. coli* (ATCC 13706™) and pre-warmed Nutrient Soft Agar, and this mixture was poured onto Nutrient Agar plates. Plates were incubated overnight at 37°C and plaques were counted the following day.

Protein concentrations were determined using the Pierce™ 660 nm Protein Assay and the BSA and bovine gamma globulin (BGG) standard pre-diluted sets to produce the calibration curves used for quantitation. Samples were diluted to give concentrations within the calibration curve, then mixed with the assay reagent. Protein concentrations were measured on a Cary-60 UV-Vis spectrophotometer.

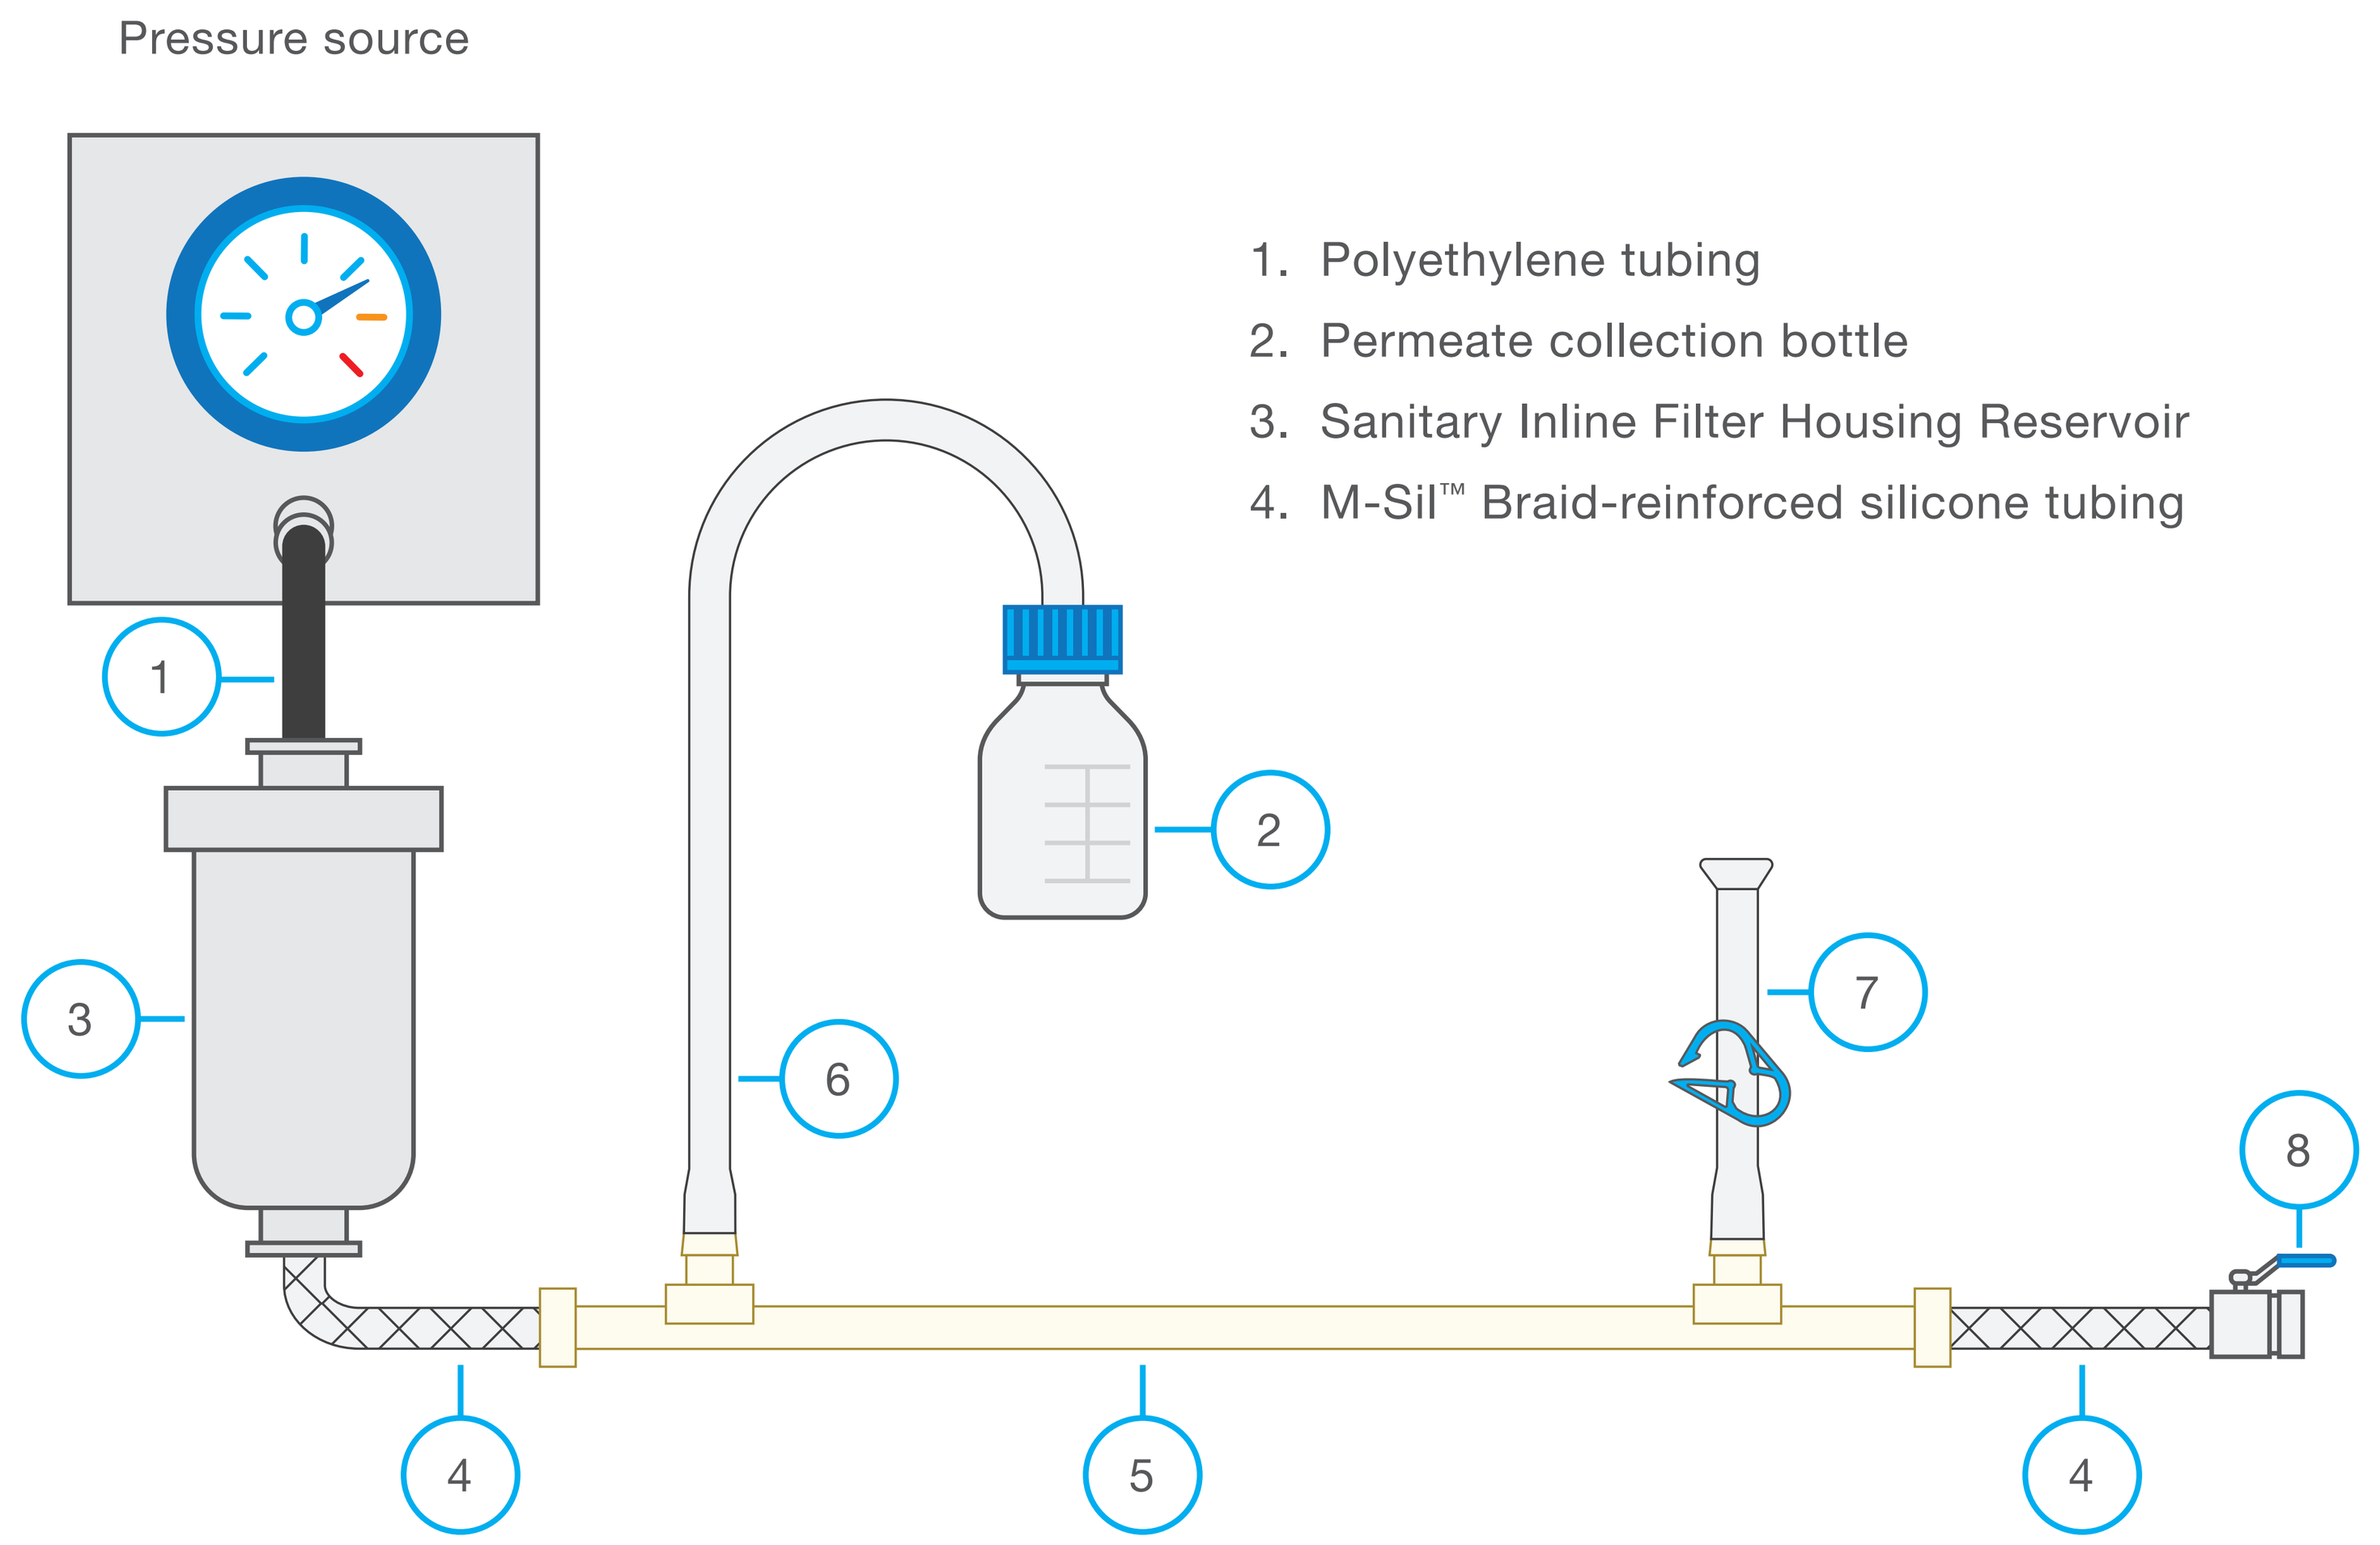
Pressure source
1
2
3
4
5
6
7
8
4

1. Polyethylene tubing
2. Permeate collection bottle
3. Sanitary Inline Filter Housing Reservoir
4. M-Sil™ Braid-reinforced silicone tubing
5. UF filter
6. Silicone tubing
7. Silicone tubing, clamped
8. Retentate Valve

Feed Concentration

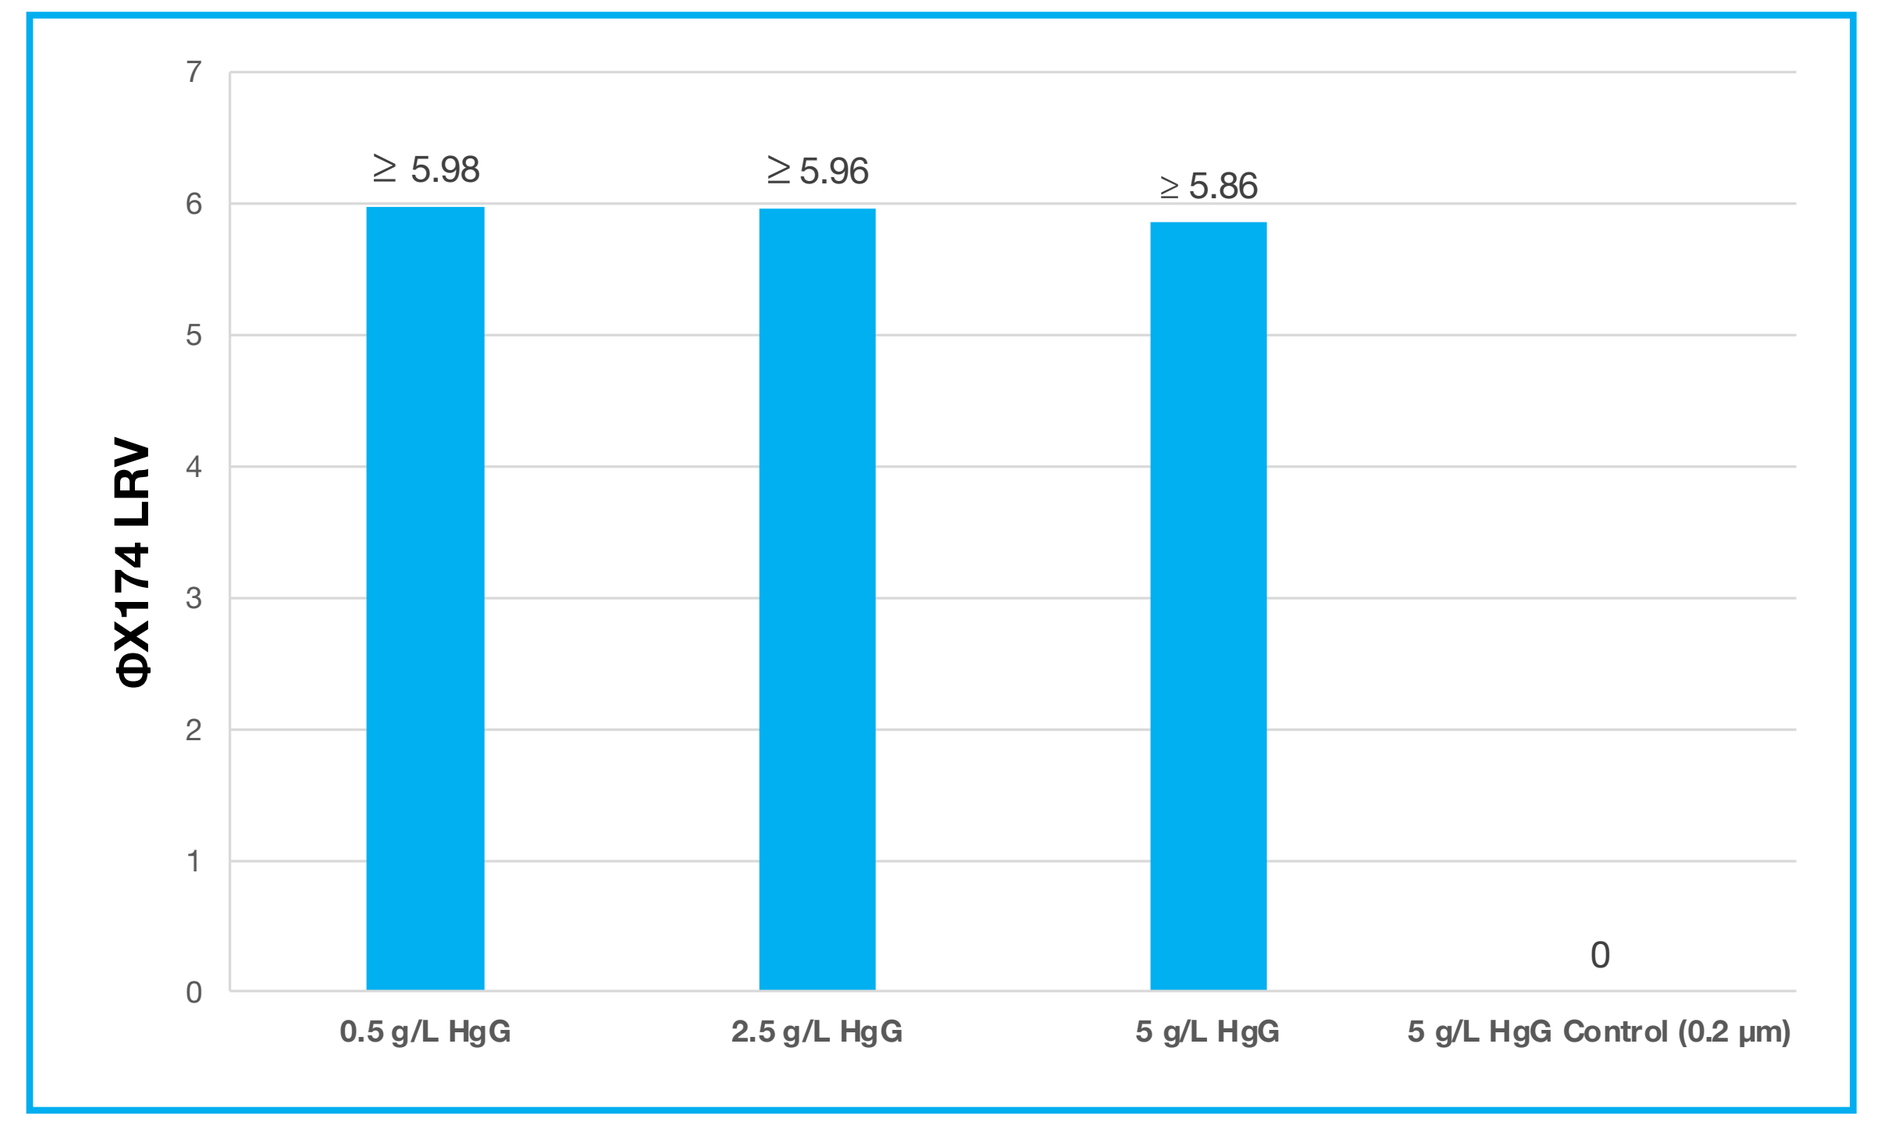
ΦX174 LRV
≥ 5.98
≥ 5.96
≥ 5.86
0
0.5 g/L HgG
2.5 g/L HgG
5 g/L HgG
5 g/L HgG Control (0.2 µm)

Figure 1: Log reduction values (LRV) of ΦX174 phage particles suspended in various concentrations of HGG

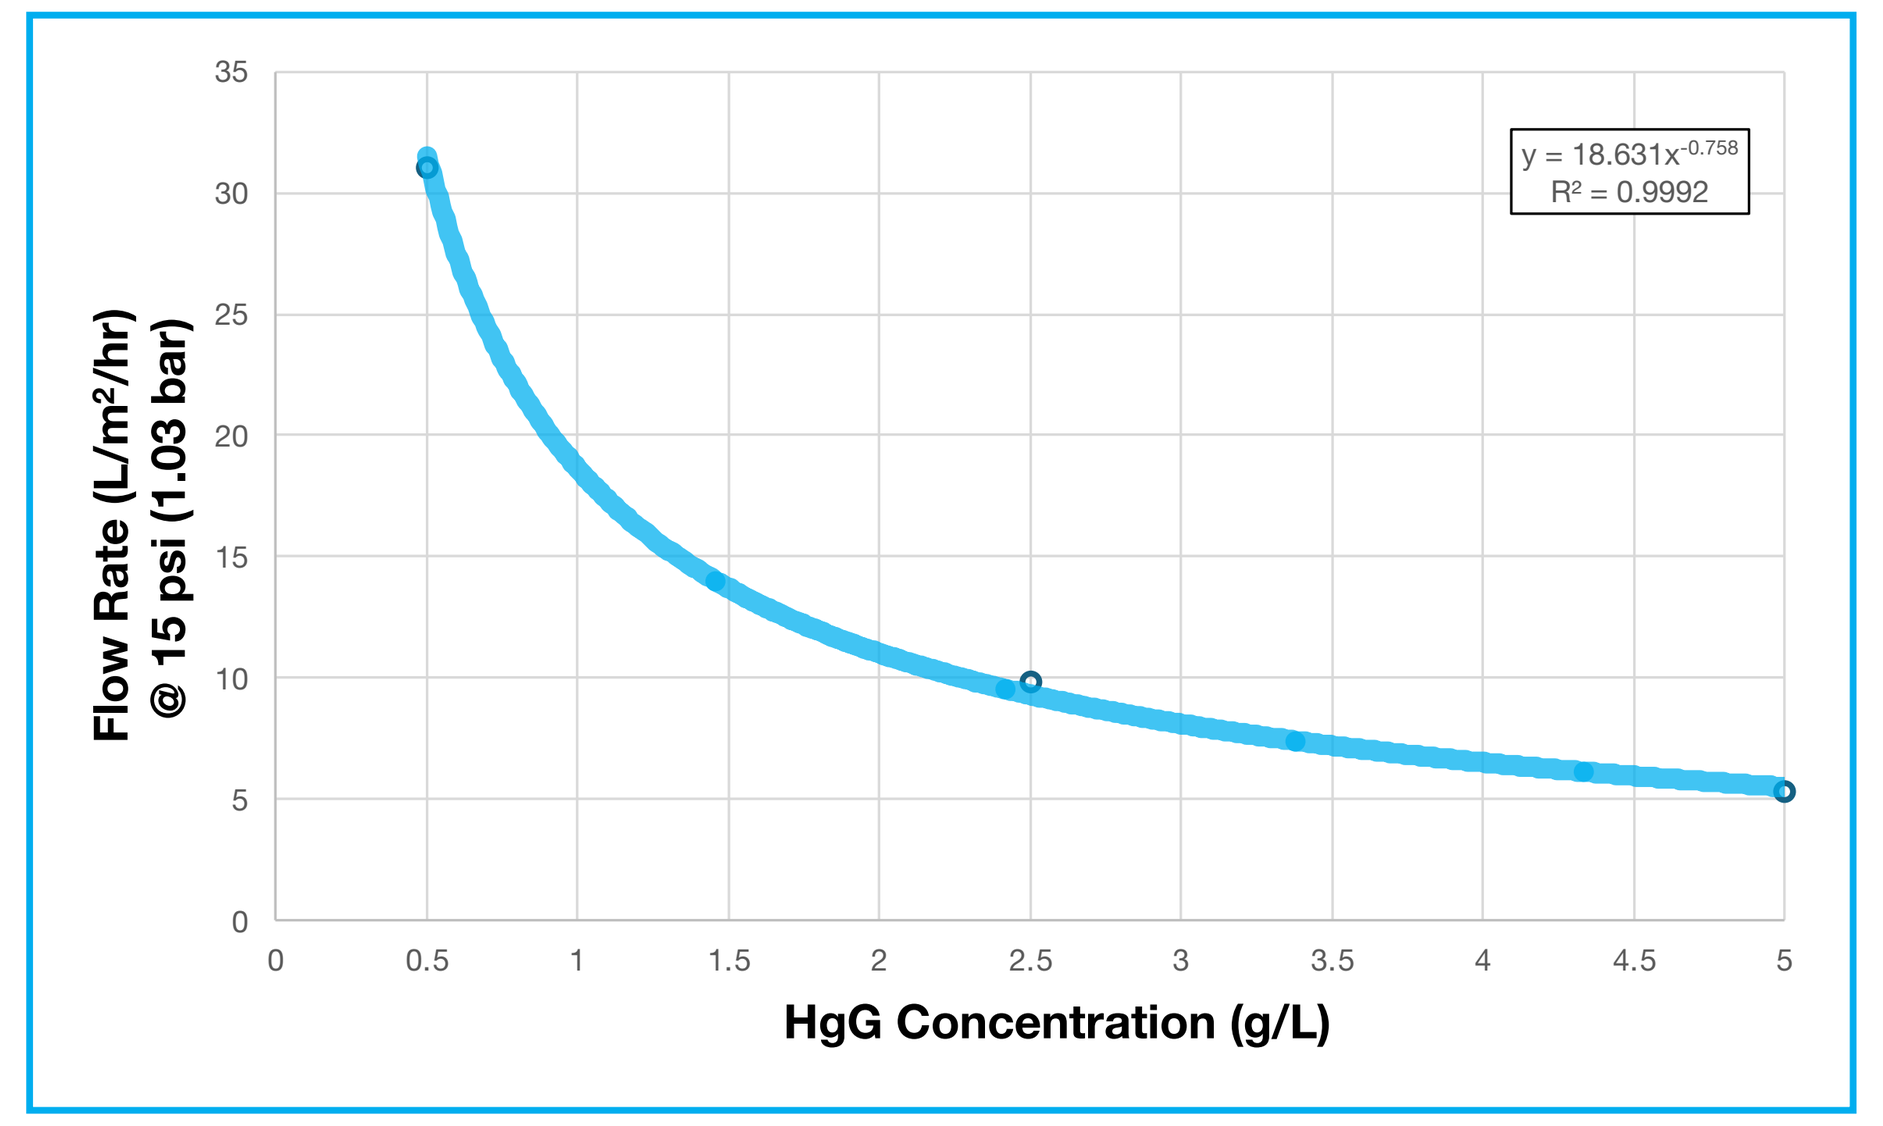
Flow Rate (L/m²/hr) @ 15 psi (1.03 bar)
y = 18.631x^-0.758
R² = 0.9992
HgG Concentration (g/L)

Figure 2: Flow rates of HgG solutions at various concentrations at 15 psi (1.03 bar) constant pressure

Protein Type

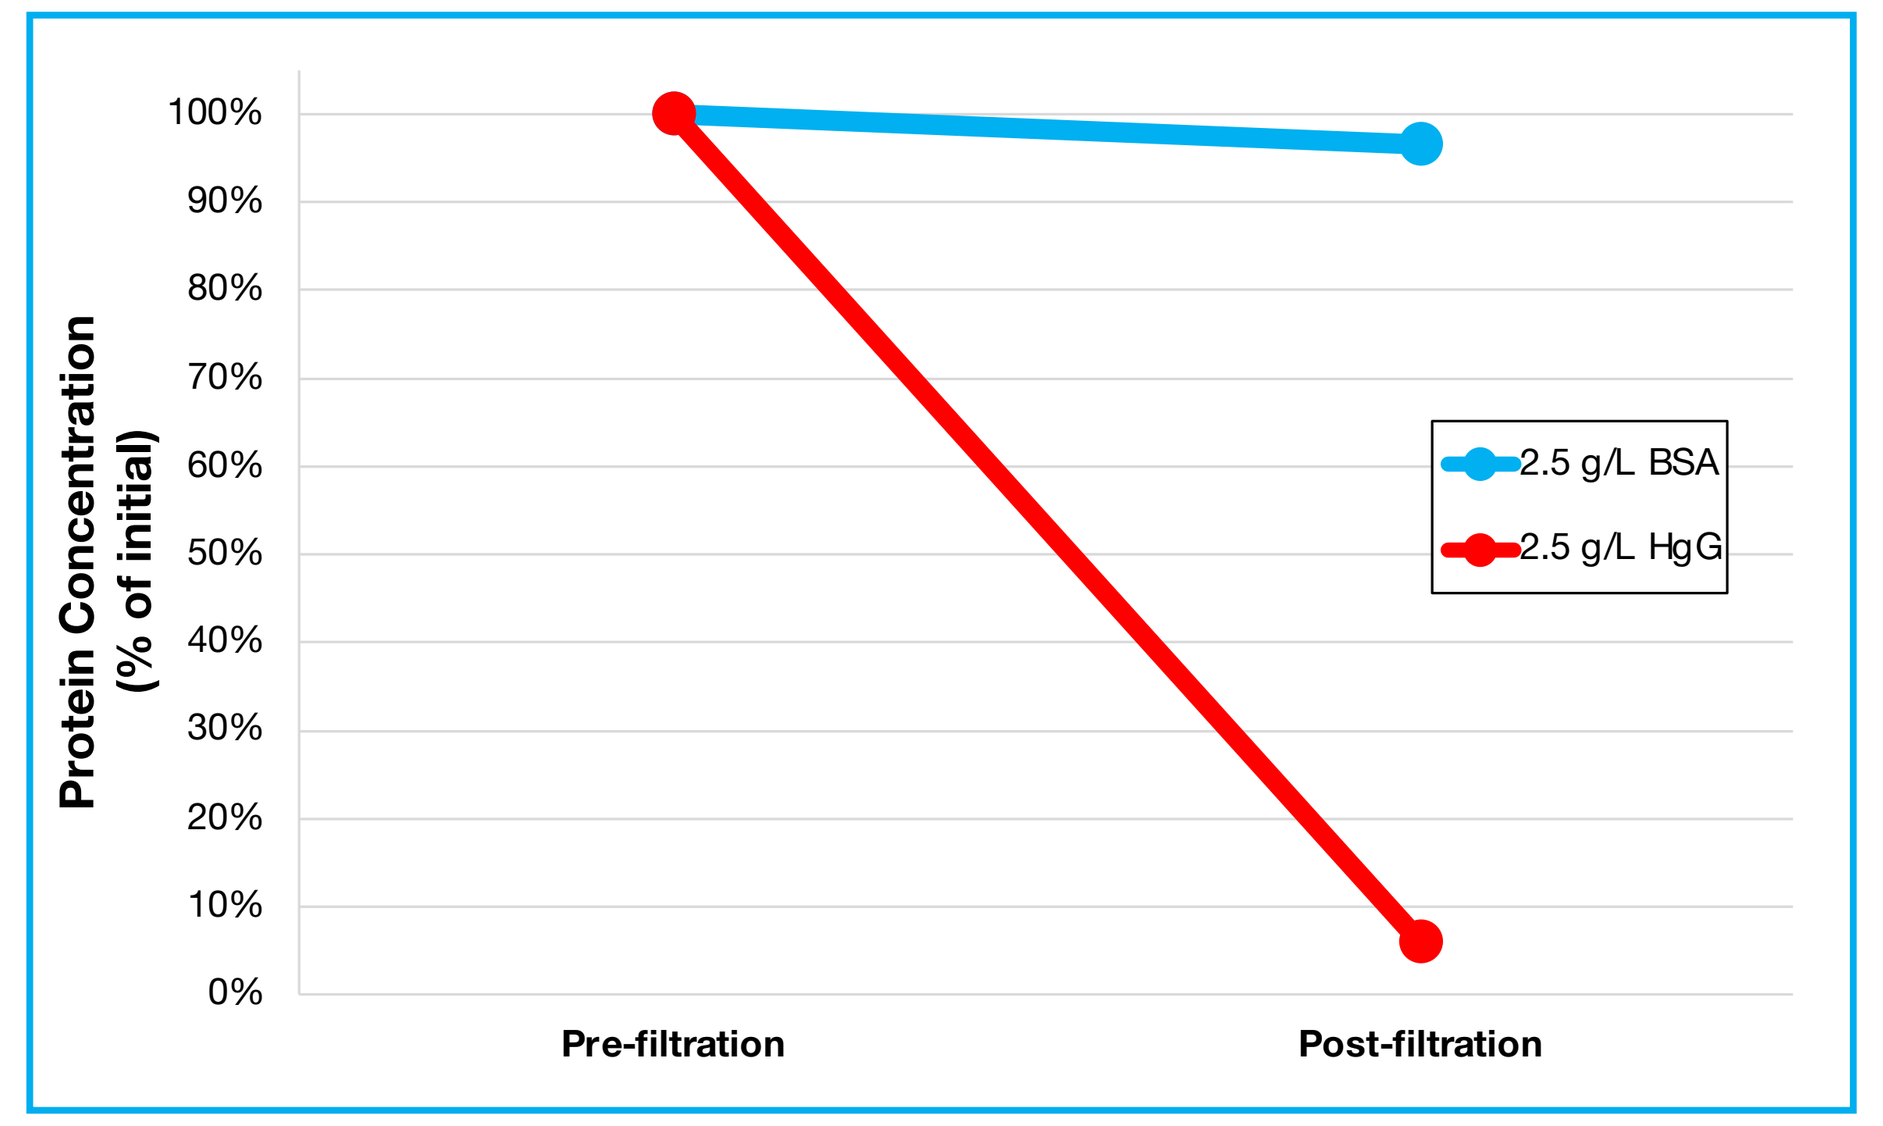
Protein Concentration (% of initial)
2.5 g/L BSA
2.5 g/L HgG
Pre-filtration
Post-filtration

Figure 3: BSA and HgG percent recovery pre- and post-filtration 15 psi (1.03 bar) constant pressure

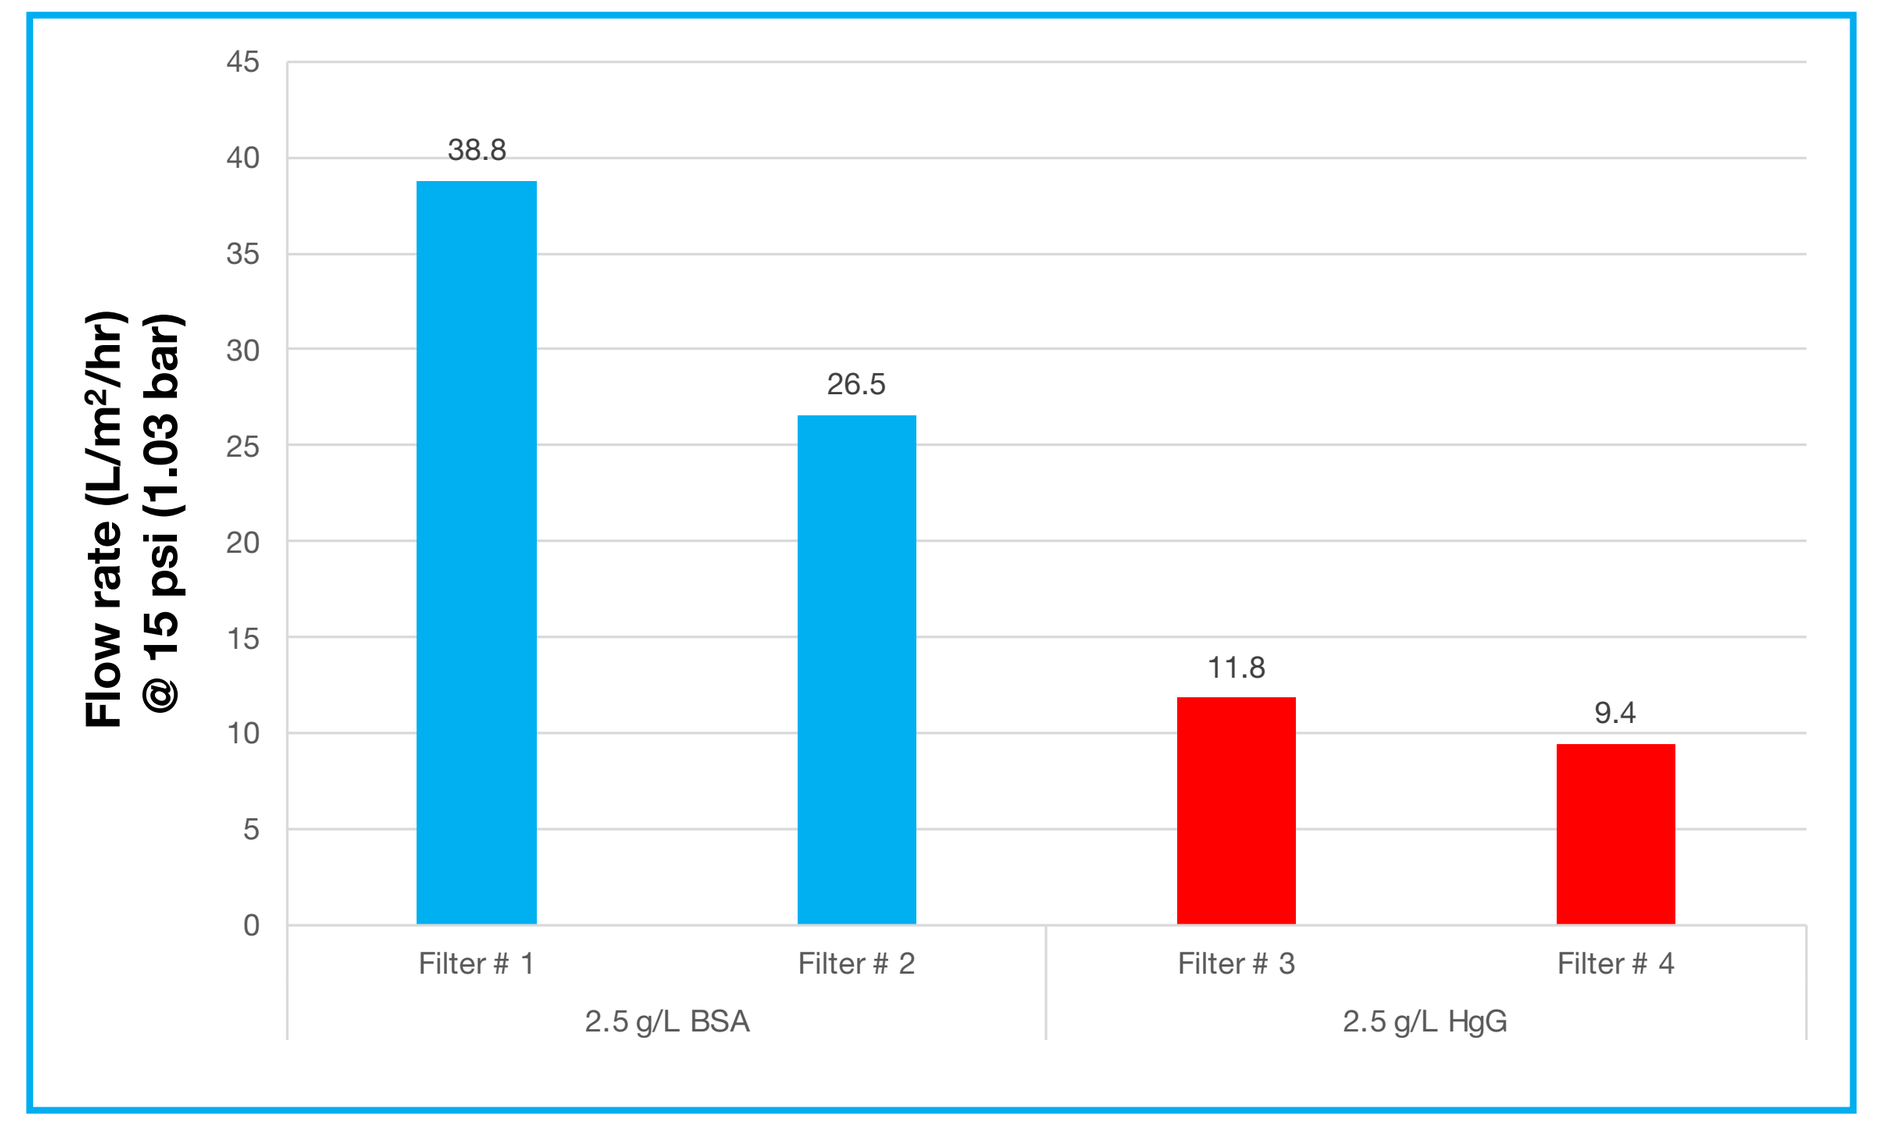
Flow rate (L/m²/hr) @ 15 psi (1.03 bar)
38.8
26.5
11.8
9.4
Filter # 1
Filter # 2
Filter # 3
Filter # 4
2.5 g/L BSA
2.5 g/L HgG

Figure 4: 2.5 g/L BSA and 2.5 g/L HGG flow rates at 15 psi (1.03 bar) constant pressure

Pressure

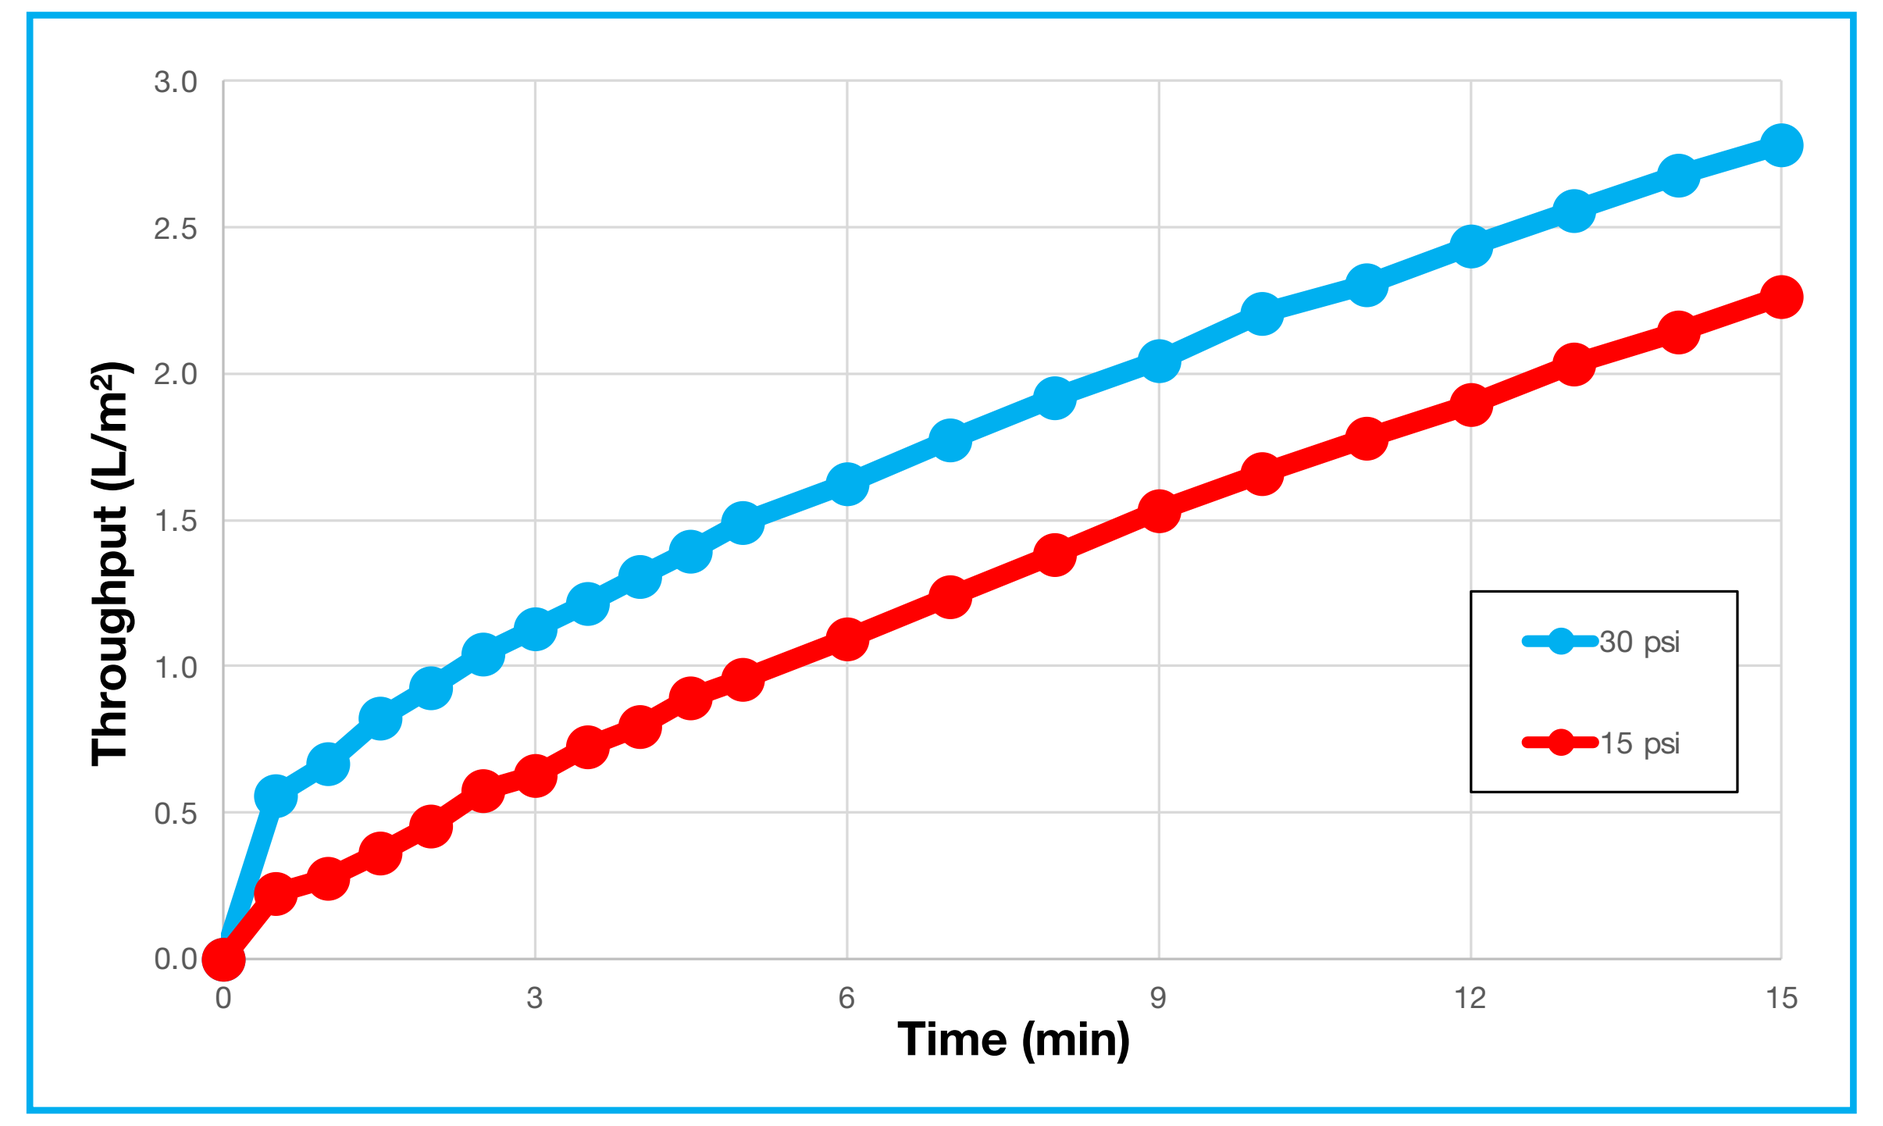
Throughput (L/m²)
30 psi
15 psi
Time (min)

Figure 5: Throughput over time for 2.5 g/L HgG solution at 15 psi (1.03 bar) and 30 psi (2.07 bar) constant pressure

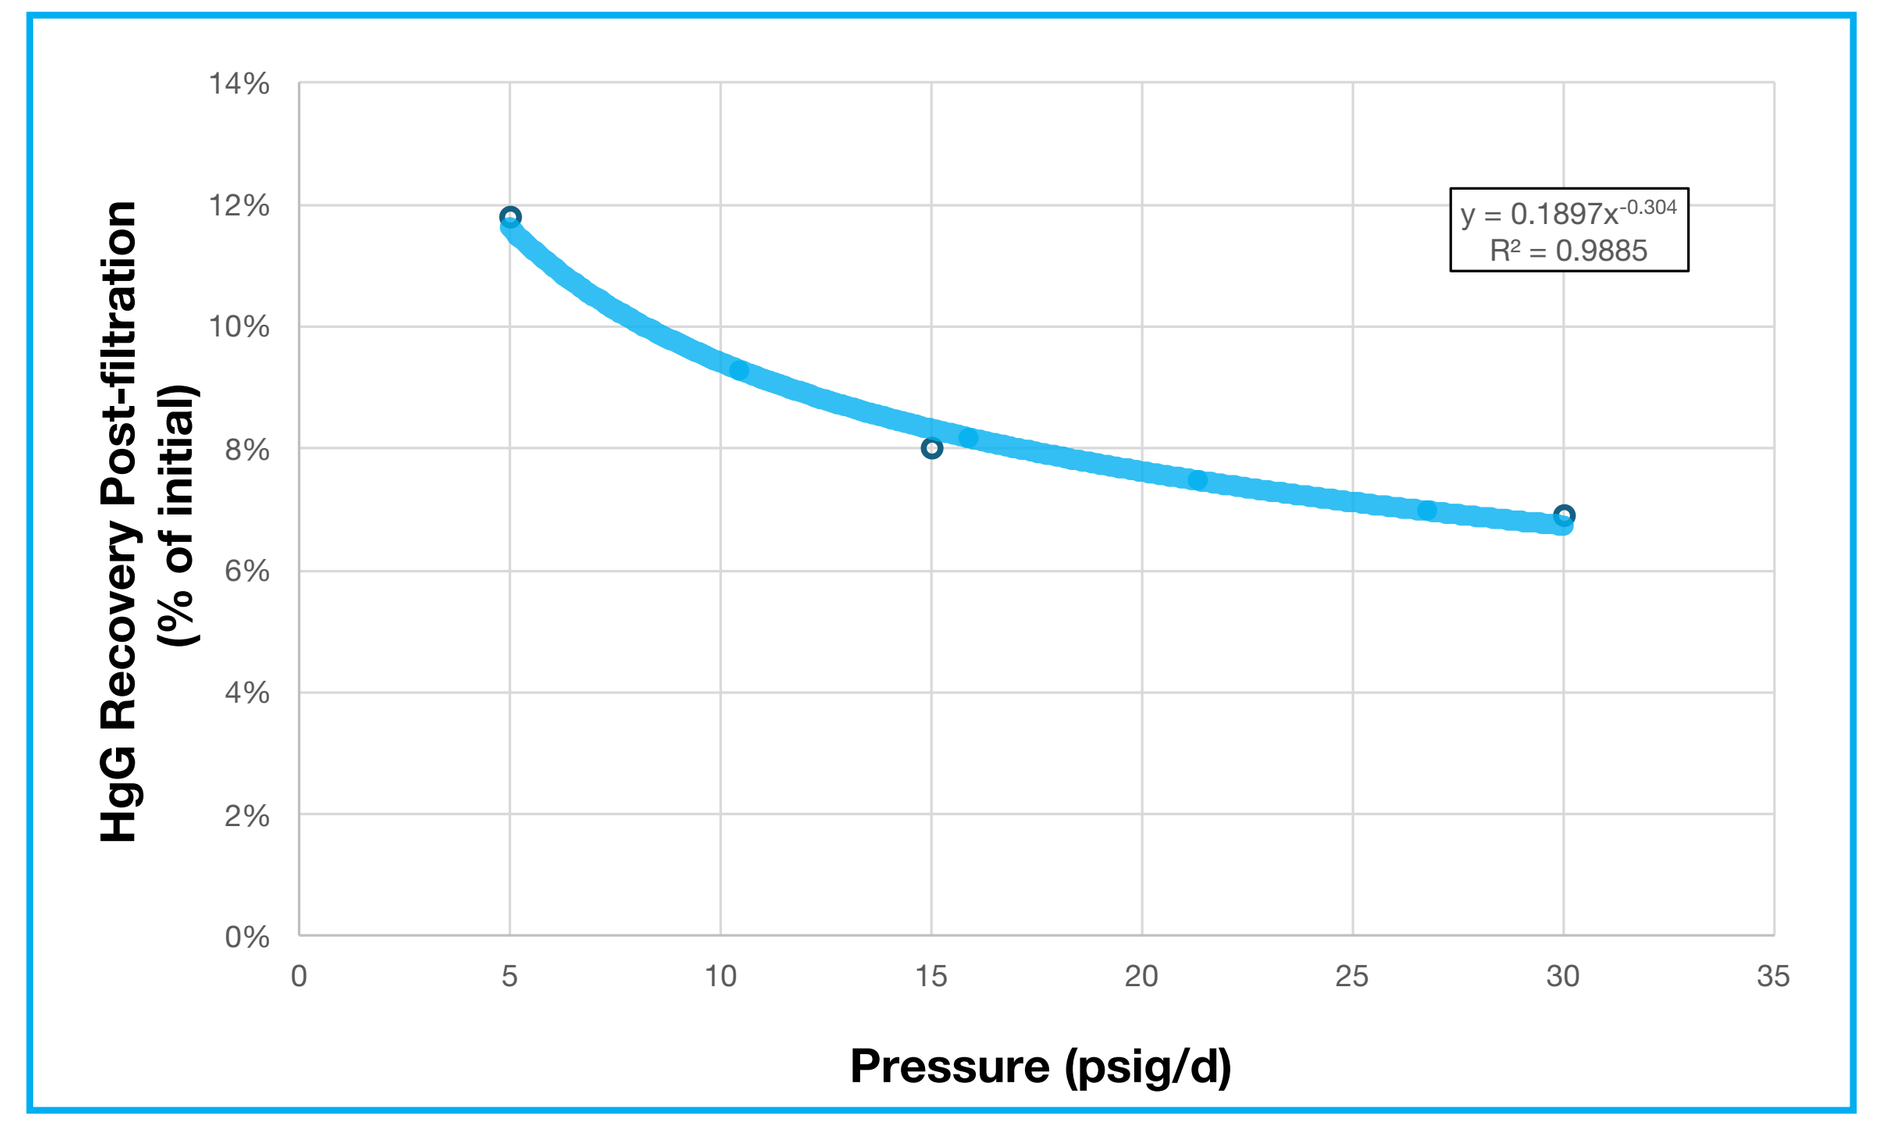
HgG Recovery Post-filtration (% of initial)
y = 0.1897x^-0.304
R² = 0.9885
Pressure (psig/d)

Figure 6: HgG percent recovery pre- and post-filtration at 5 to 30 psi (0.34 to 2.07 bar) constant pressure

Conclusion

In this study, three key parameters (feed concentration, protein size, and inlet pressure) were examined for their impact on viral retention, protein passage, and flow rates during ultrafiltration. Feed concentration, protein size, and inlet pressure were all found to influence product flow rates. Inlet pressure and the MW of the protein filtered were shown to affect downstream protein recovery. However, feed concentration was not found to significantly affect viral retention. This work demonstrates the complex interplay of factors that influence ultrafiltration performance and will help end-users design process parameters that maximize efficiency while minimizing negative impacts to product quality and safety.

www.meissner.com
For additional information, please contact: info@meissner.com

MEISSNER